Anti-DR5 agonist IgM antibody IGM-8444 combined with SMAC mimetic birinapant induces strong synergistic tumor cytotoxicity

Beatrice T. Wang, Mélanie Desbois, Susan E. Calhoun, Thomas J. Matthew, Poonam Yakkundi, Ling Wang, Xingjie Chen, Tasnim Kothambawala, Miho Oyasu, Maya F. Kotturi, Genevive Hernandez, Xiaohan Liu, Marvin S. Peterson, Eric W. Humke, Bruce A. Keyt, Angus M. Sinclair

IGM Biosciences, Inc. | Mountain View, CA

Background

- IGM-8444 is a multivalent IgM agonist that targets tumor necrosis factor (TNF) receptor superfamily member death receptor DR5, which requires multimerization to induce tumor cell apoptosis. IGM-8444 is currently being evaluated in a first-in-human Phase 1 trial.
- Birinapant is a mimetic of endogenous second mitochondria-derived activator of caspases (SMAC) that blocks inhibitor of apoptosis proteins (IAPs) and has been evaluated through Phase 2, demonstrating good safety and on target activity but minimal efficacy as a single agent.
- Since apoptosis is regulated through both extrinsic and intrinsic signaling pathways, we hypothesized that simultaneously targeting the extrinsic apoptotic pathway with IGM-8444 and the intrinsic apoptotic pathway with birinapant could overcome cellular resistance mechanisms and enhance tumor cytotoxicity.

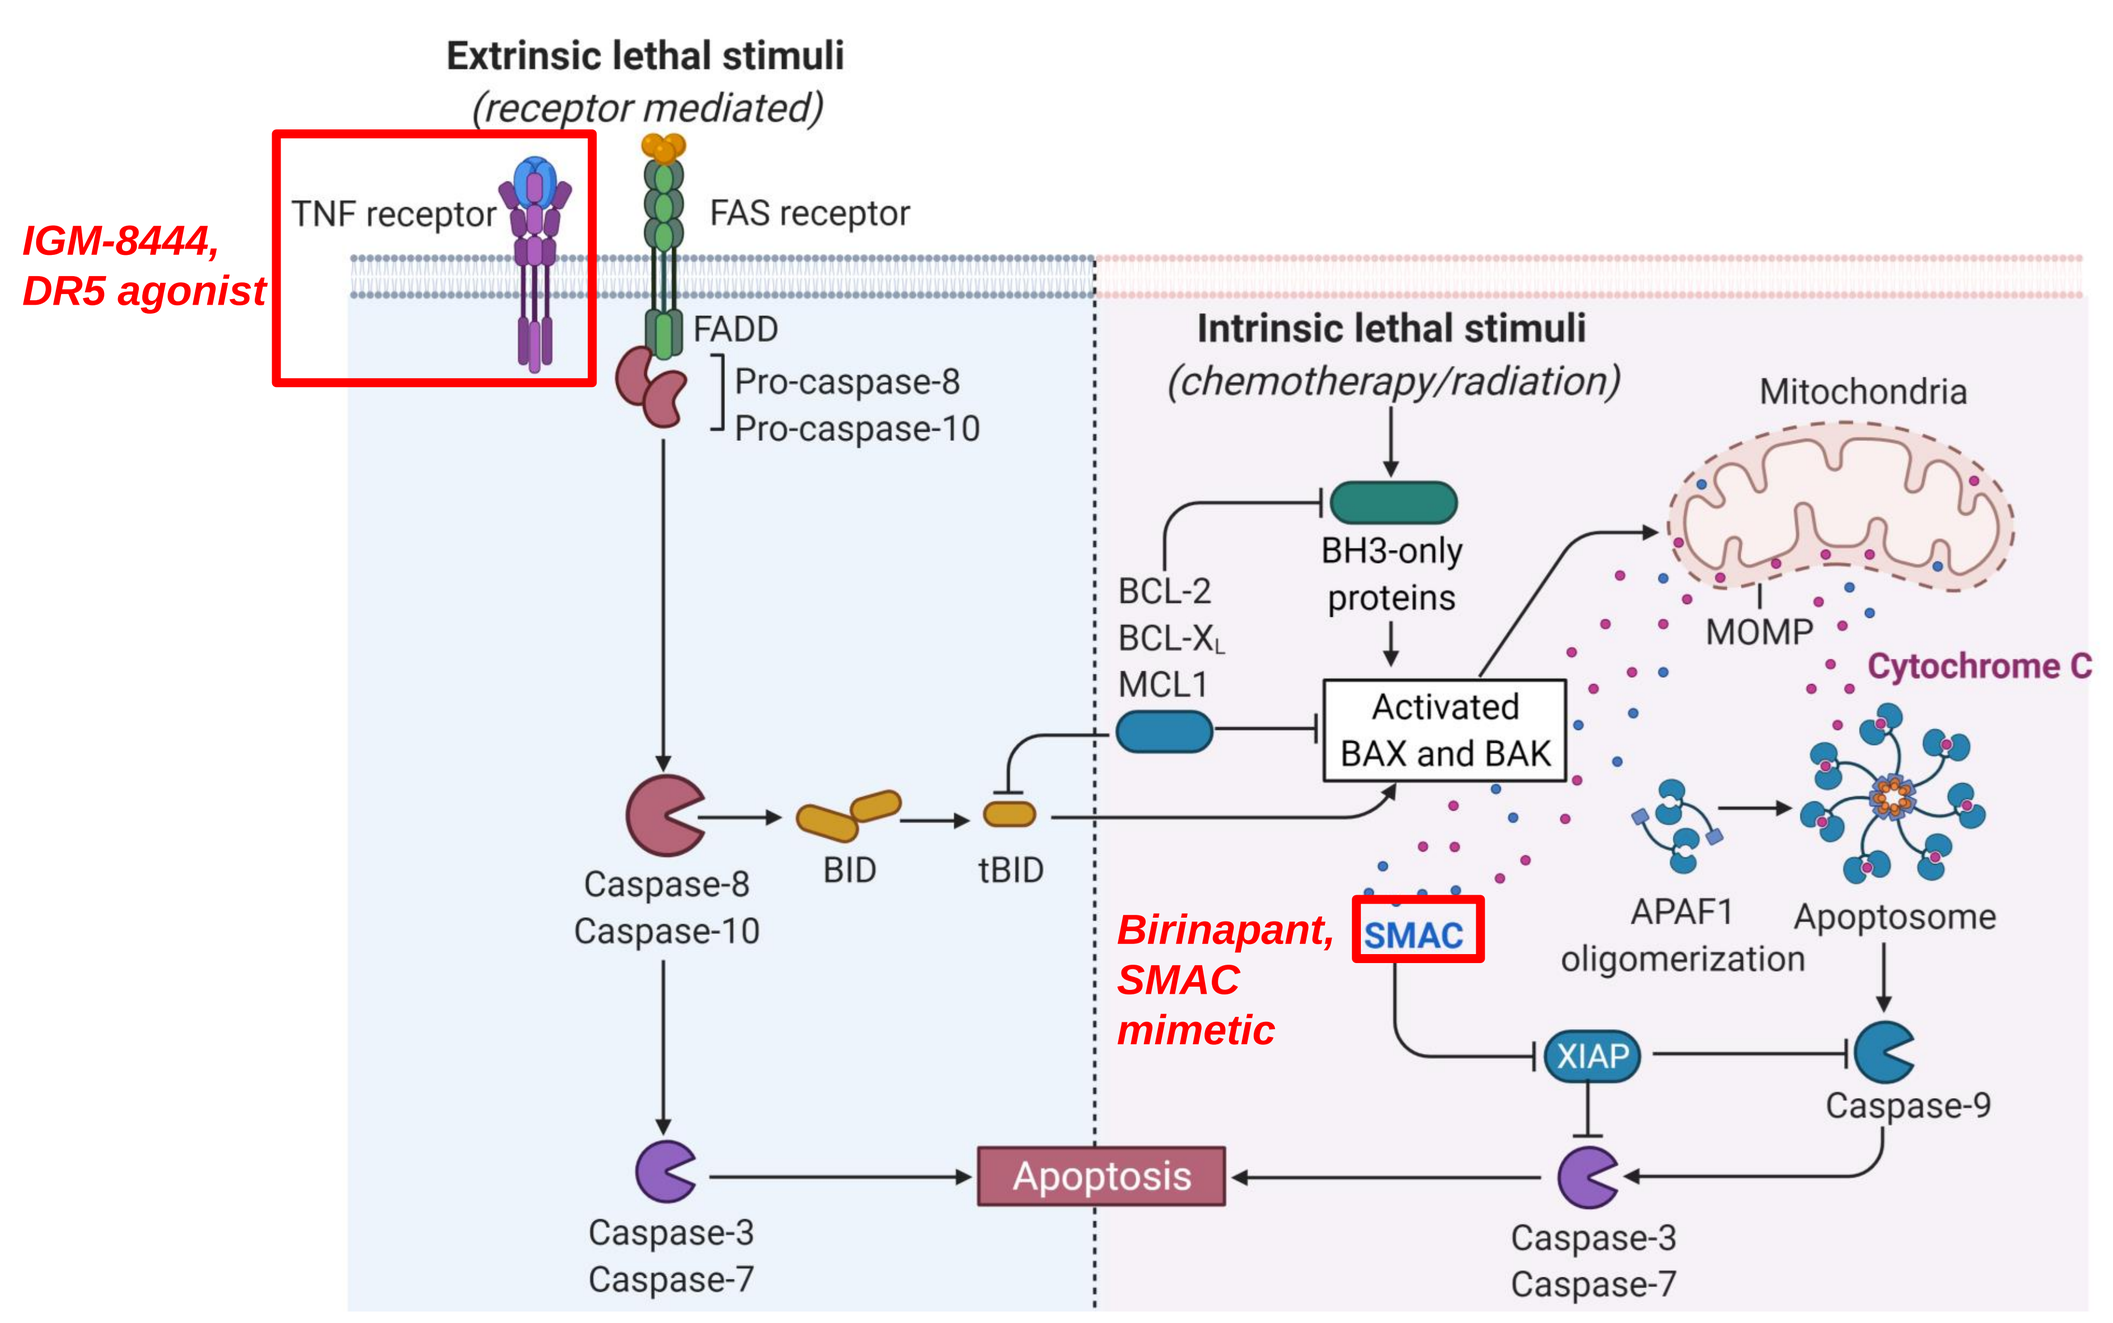

Extrinsic lethal stimuli
(receptor mediated)
IGM-8444, DR5 agonist
TNF receptor
FAS receptor
FADD
Pro-caspase-8
Pro-caspase-10
Intrinsic lethal stimuli
(chemotherapy/radiation)
Mitochondria
BH3-only proteins
BCL-2
BCL-XL
MCL1
MOMP
Cytochrome C
Activated BAX and BAK
Caspase-8
Caspase-10
BID
tBID
Birinapant, SMAC mimetic
SMAC
APAF1 oligomerization
Apoptosome
XIAP
Caspase-9
Caspase-3
Caspase-7
Apoptosis

Figure 1. Extrinsic and intrinsic apoptotic signaling pathways. Created with BioRender.com.

IGM-8444 and Birinapant Combination Induced Synergistic Cytotoxicity Across Multiple Human Cancer Cell Lines

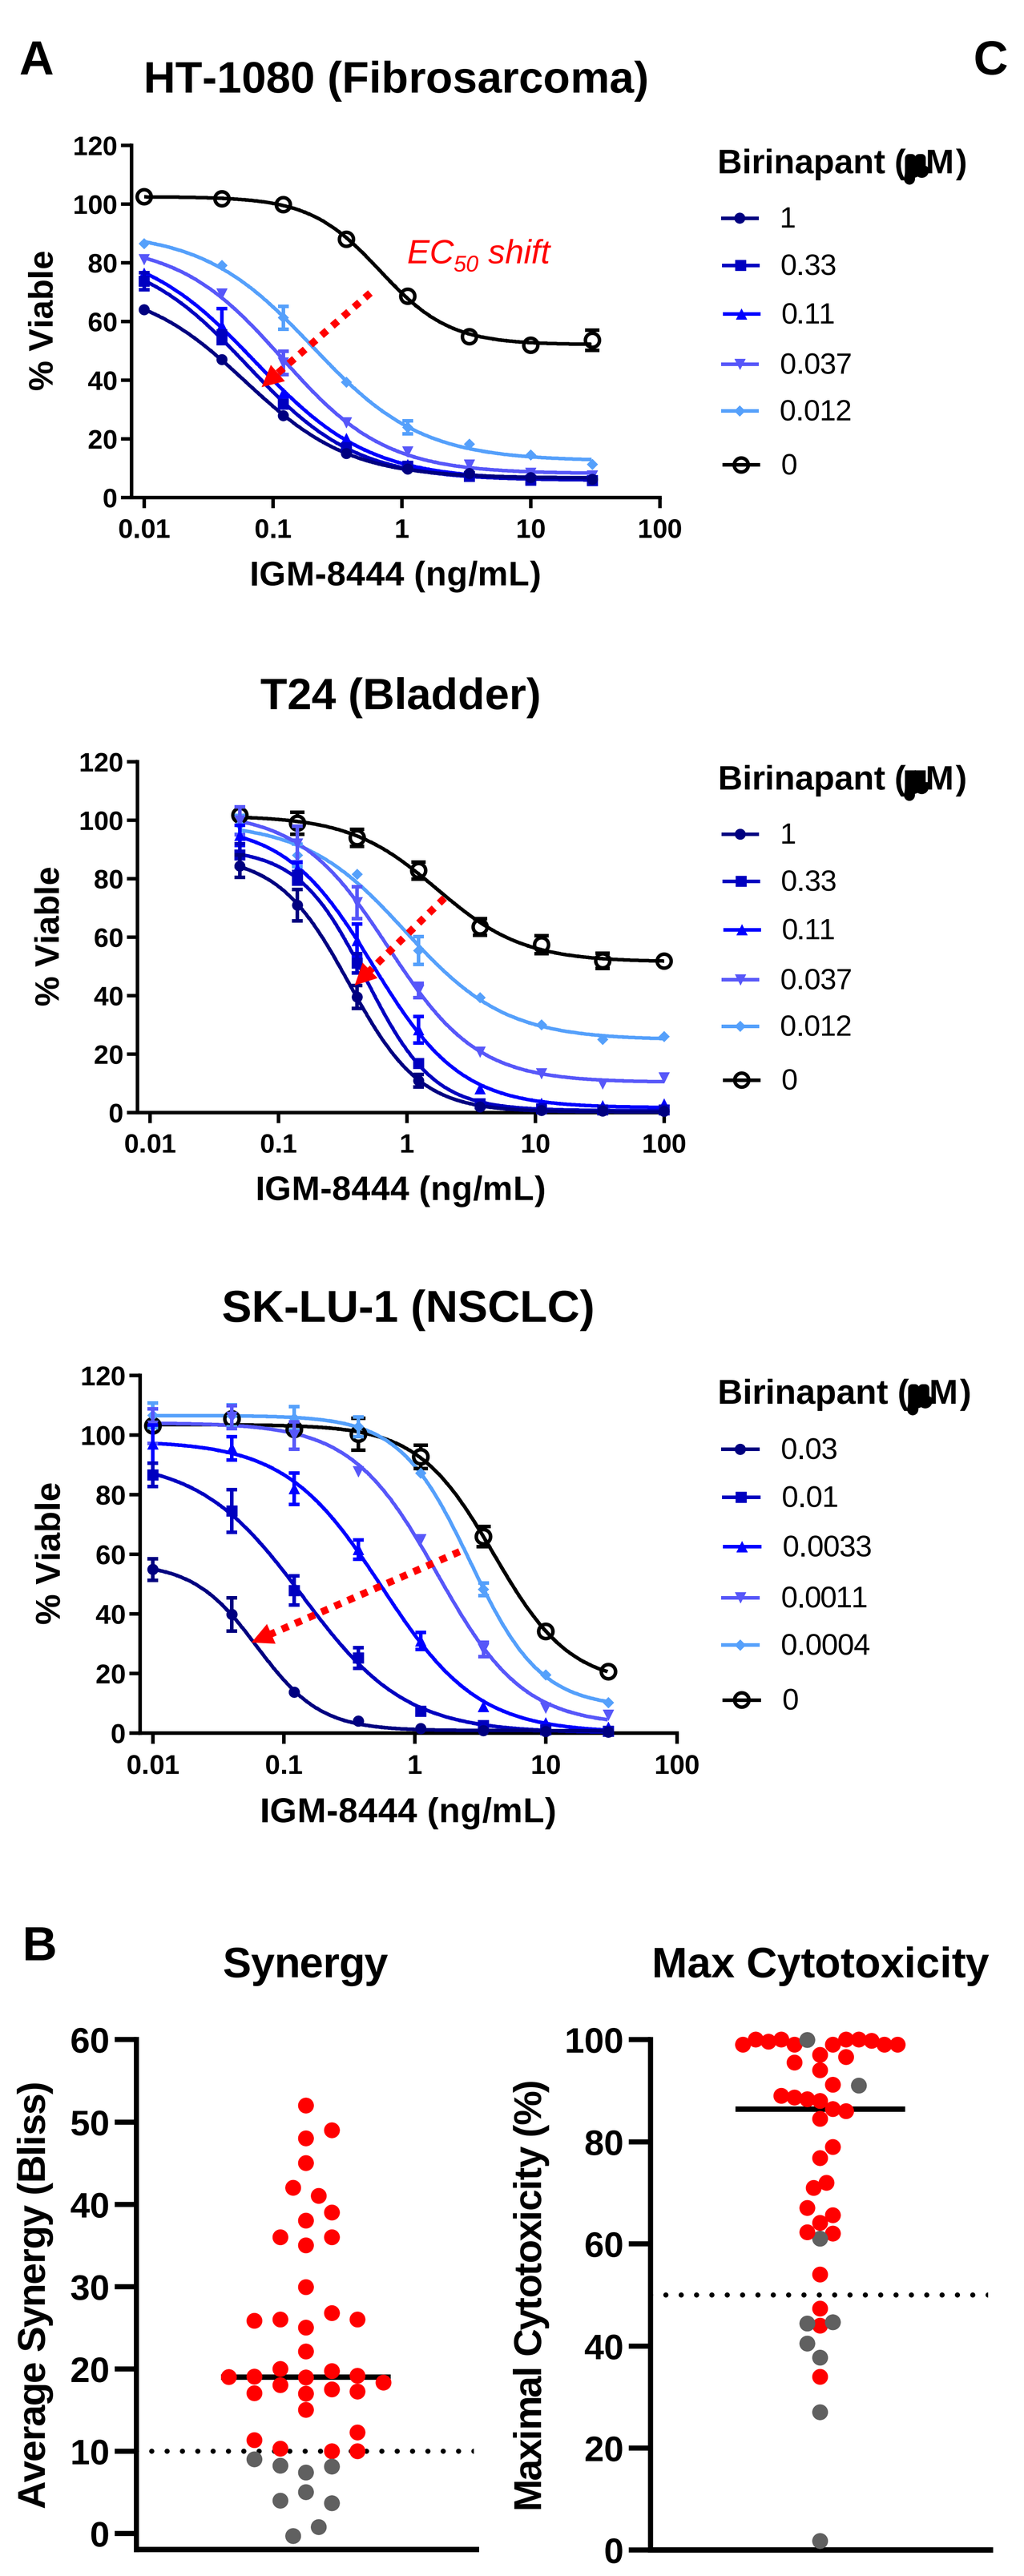

A
HT-1080 (Fibrosarcoma)
EC50 shift
% Viable
IGM-8444 (ng/mL)
Birinapant (μM)
1
0.33
0.11
0.037
0.012
0
T24 (Bladder)
SK-LU-1 (NSCLC)
0.03
0.01
0.0033
0.0011
0.0004
B
Synergy
Max Cytotoxicity
Average Synergy (Bliss)
Maximal Cytotoxicity (%)

C

Human Cell Line	Tumor Type	Average Bliss Synergy Score
MDA-MB-231	TNBC	52
A2058	Melanoma	49
EBC-1	Lung (SCC)	48
HT 1080	Fibrosarcoma	45
KYSE 410	Esophageal	42
HCC15	Lung (SCC)	41
SNU-1	Gastric	39
T24	Bladder	38
HCT 116	Colorectal	36
CCK-81	Colorectal	36
SW780	Bladder	35
SW579	Head & Neck	30
DETROIT562	Head & Neck	27
Saos-2	Osteosarcoma	26
MEWO	Melanoma	26
KYSE410	Head & Neck	26
SK-LU-1	Lung (adeno)	25
TE1	Head & Neck	22
OVCAR-4	Ovarian	20
FTC133	Head & Neck	20
8305C	Head & Neck	19
SCC25	Head & Neck	19
ES-2	Ovarian	19
A253	Head & Neck	19
KYSE70	Head & Neck	18
SW1353	Chondrosarcoma	18
SCC9	Head & Neck	17
BCPAP	Head & Neck	17
CAL62	Head & Neck	17
OVCAR-5	Ovarian	17
SK-MEL 5	Melanoma	15
OE19	Head & Neck	12
FADU	Head & Neck	11
KYSE270	Head & Neck	10
KP 4	Pancreatic	10
SK-ES-1	Sarcoma	10
DV-90	Lung (adeno)	9
SCC15	Head & Neck	8
KMH2	Head & Neck	8
8505C	Head & Neck	7
SCC4	Head & Neck	5
BT-20	TNBC	4
CAL27	Head & Neck	4
KYSE150	Head & Neck	0.8
T84	Colorectal	-0.3

Figure 2. Human cell lines across multiple solid tumor indications (n=45) were treated with IGM-8444 and birinapant and viability was measured after 72 hours using CellTiter-Glo. **A)** Representative cell lines are shown. Dotted red arrow indicates EC50 shift, suggestive of synergy. **B)** Synergy was calculated using Bliss scores, where a Bliss score greater than 10 indicates synergy. 80% of cell lines (36 out of 45, red dots) had average Bliss scores greater than 10 (median = 19, maximum = 52), and 80% of cell lines (36 out of 45, red dots) showed greater than 50% maximal killing (median = 86%). **C)** List of cell lines tested and associated average Bliss scores. Synergistic Bliss scores are highlighted in teal.

IGM-8444 and Birinapant Combination Induced Synergistic Killing in Cell Line with Acquired Resistance to DR5 Agonists

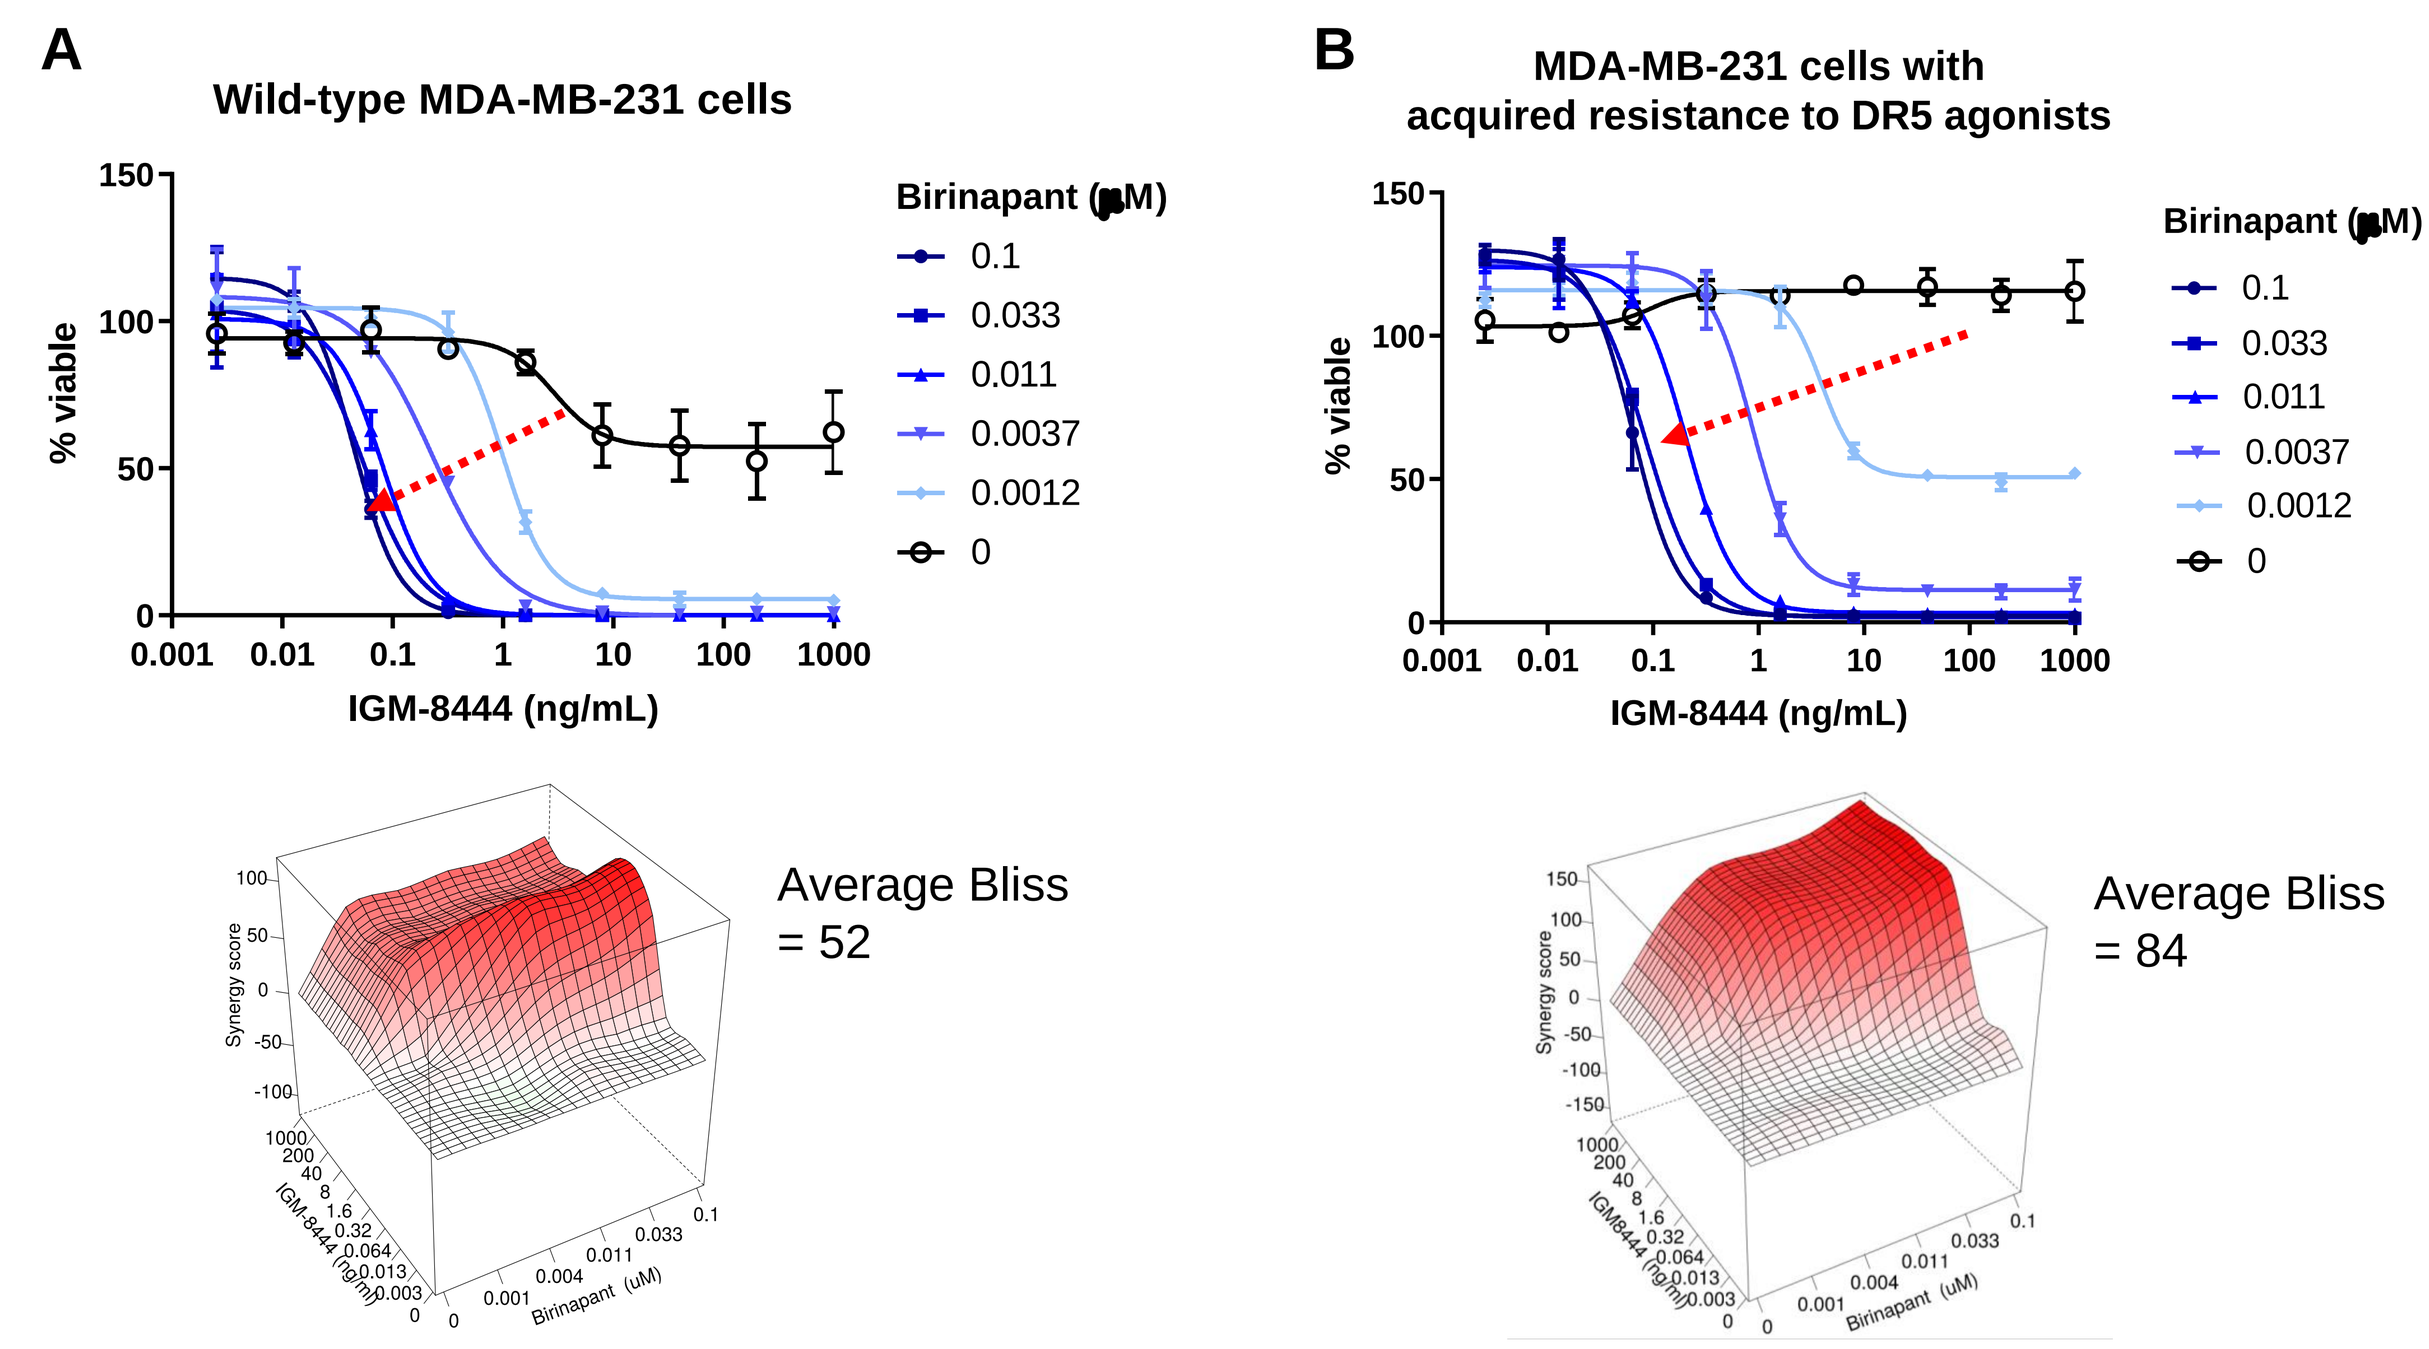

A
Wild-type MDA-MB-231 cells
B
MDA-MB-231 cells with acquired resistance to DR5 agonists
% viable
IGM-8444 (ng/mL)
Birinapant (μM)
0.1
0.033
0.011
0.0037
0.0012
0
Average Bliss = 52
Average Bliss = 84
Synergy score
Birinapant (uM)

Figure 3. MDA-MB-231 cells with acquired resistance to a DR5 agonist were generated by culturing wild-type MDA-MB-231 cells in the presence of 0.1 μg/mL of an anti-DR5 IgM, and resistance to DR5 agonists including IGM-8444 was confirmed in a cytotoxicity assay. Wild-type **(A)** and acquired resistant **(B)** MDA-MB-231 cells were treated with the combination of IGM-8444 and birinapant, and viability was measured after 72 hours using CellTiter-Glo. Synergy scores were calculated across the continuum of doses and regions of synergy (Bliss scores >10) are shown in red. The synergistic response was especially notable in the acquired resistant cells, which displayed an average Bliss synergy score of 84.

IGM-8444 and Birinapant Combination Does Not Kill Primary Human Hepatocytes *in Vitro*

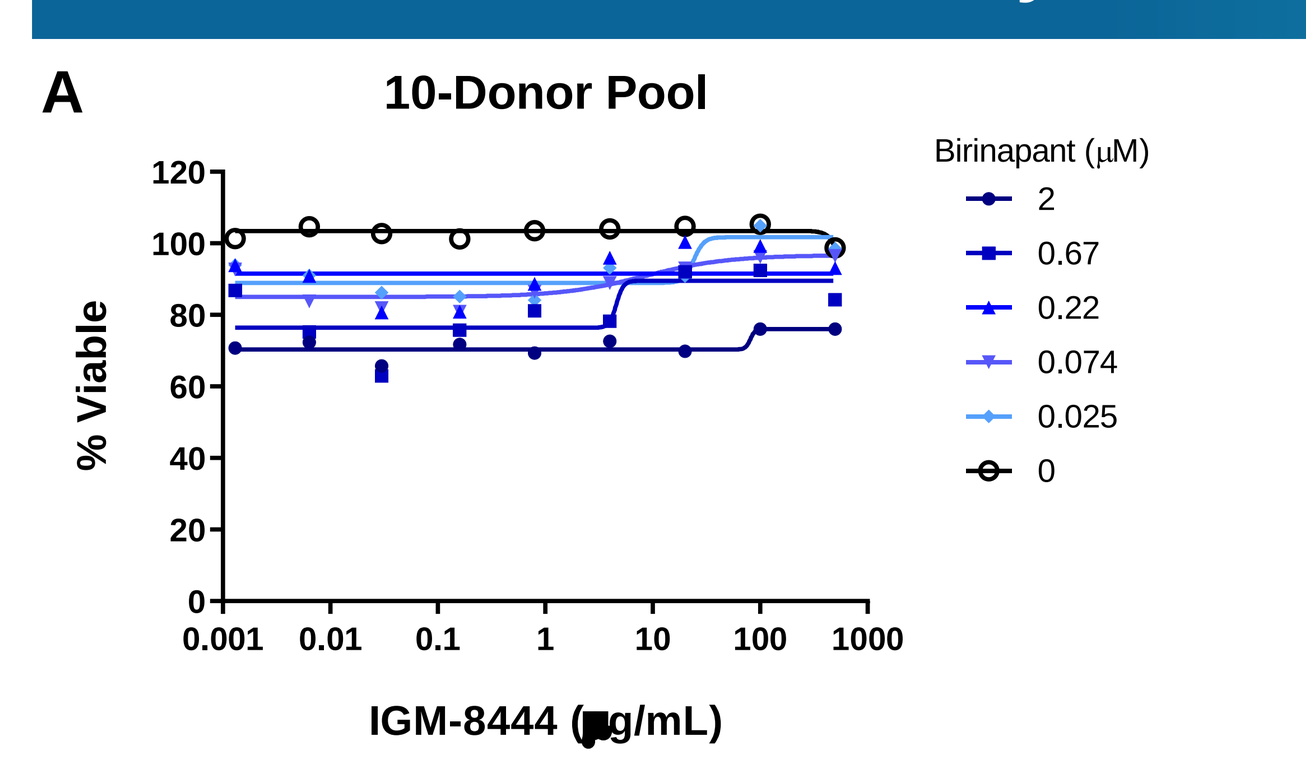

A
10-Donor Pool
% Viable
IGM-8444 (μg/mL)
Birinapant (μM)
2
0.67
0.22
0.074
0.025
0

Figure 4. Primary pooled human hepatocytes **(A)** or individual healthy donors **(B)** were treated with IGM-8444 in combination with birinapant, and viability after 72 hours was measured using CellTiter-Glo. The combined treatment did not induce cytotoxicity in primary human hepatocytes.

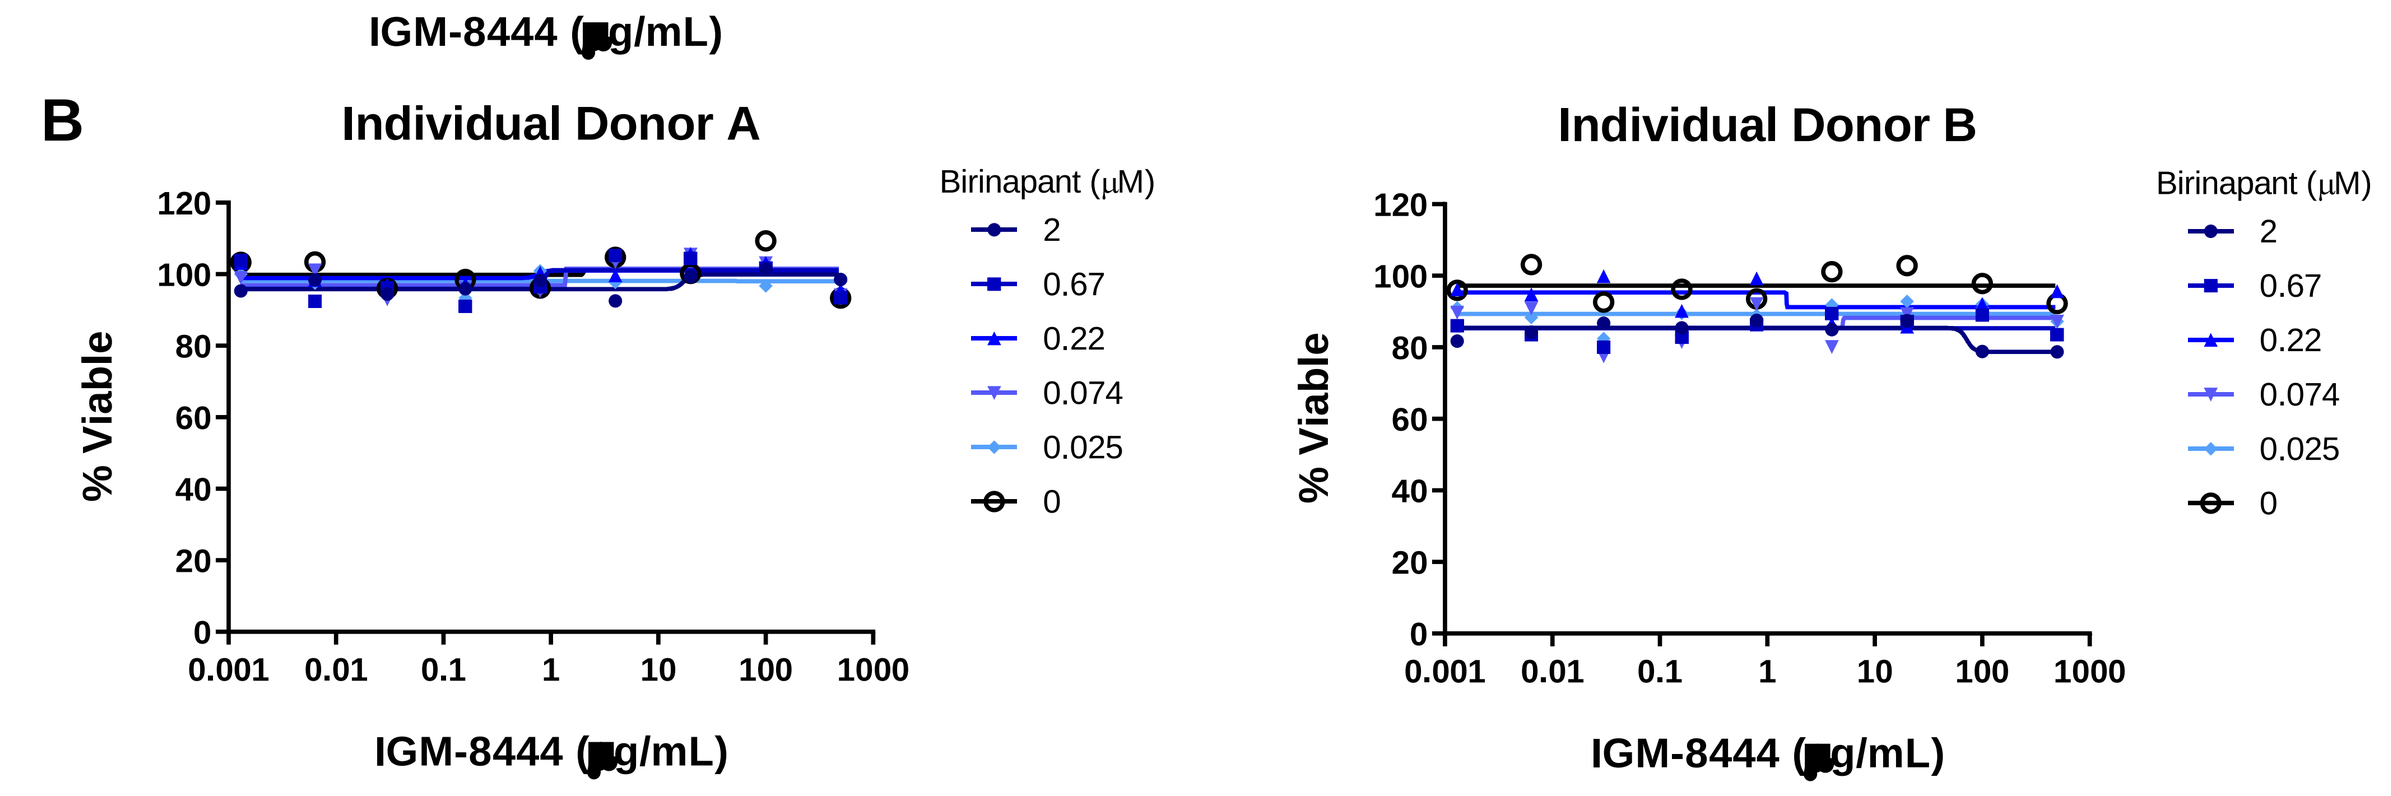

B
Individual Donor A
Individual Donor B
% Viable
IGM-8444 (μg/mL)
Birinapant (μM)
2
0.67
0.22
0.074
0.025
0

IGM-8444 and Birinapant Combination Induces Tumor Regression in Several CDX and PDX Models *In Vivo*

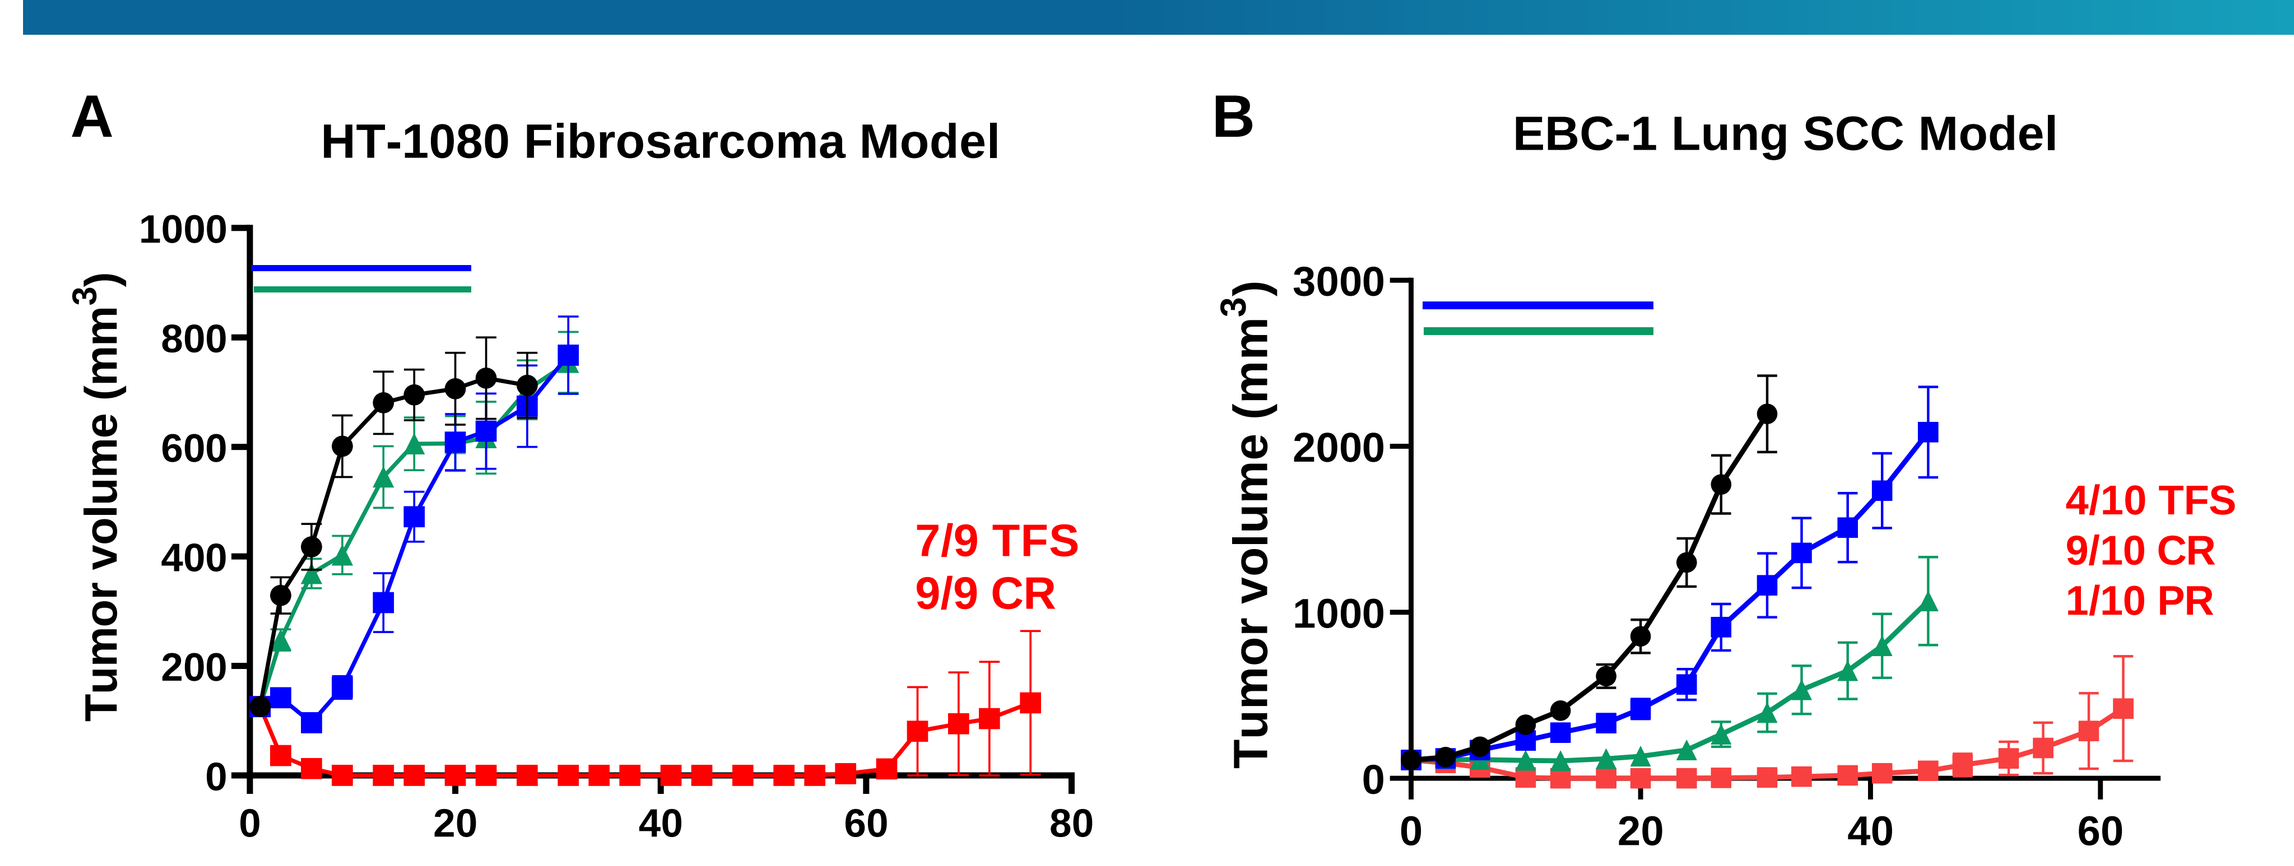

A
HT-1080 Fibrosarcoma Model
7/9 TFS
9/9 CR
B
EBC-1 Lung SCC Model
4/10 TFS
9/10 CR
1/10 PR
Tumor volume (mm3)
Days

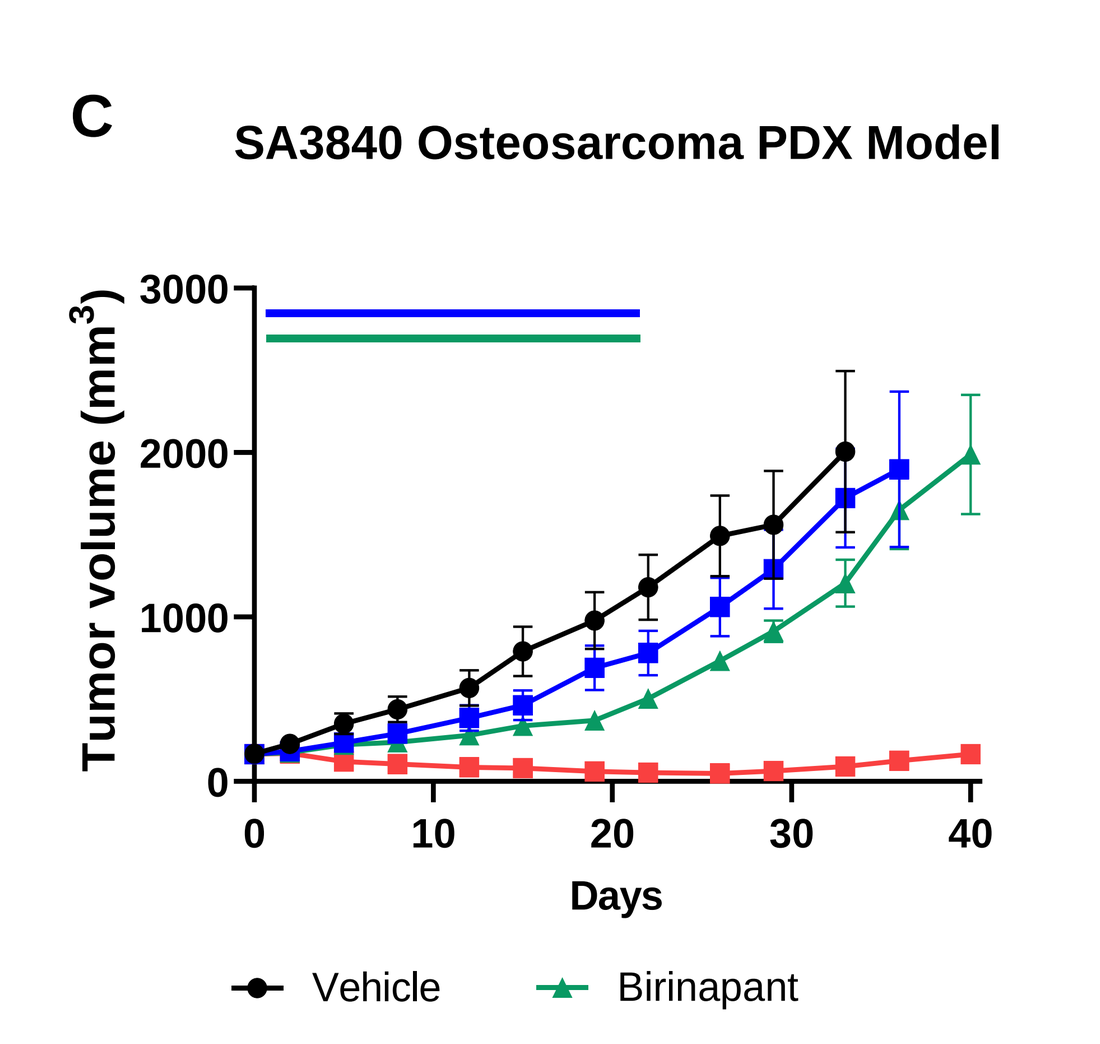

C
SA3840 Osteosarcoma PDX Model
Tumor volume (mm3)
Days
Vehicle
Birinapant
IGM-8444
IGM-8444 + Birinapant

Figure 5. A) HT-1080 fibrosarcoma cells were implanted s.c. in female athymic nude mice and dosed with vehicle i.v. q2dx11, IGM-8444 at 5 mg/kg i.v. q2dx11, birinapant at 30 mg/kg i.p. q3dx2 followed by 15 mg/kg i.p. q3dx5, or a combination of the IGM-8444 and birinapant dosing regimens. **B)** EBC-1 lung squamous cell carcinoma cells were implanted s.c. in female athymic nude mice and dosed with vehicle i.v. q2dx11, IGM-8444 at 5 mg/kg i.v. q2dx11, birinapant at 30 mg/kg i.p. q3dx7, or a combination of the IGM-8444 and birinapant dosing regimens. **C)** SA3840 osteosarcoma tumor fragments were implanted in NOD/SCID mice, then dosed as in the EBC-1 model. Tumor volumes (mean ± SEM) are plotted. Combination of IGM-8444 with birinapant resulted in enhanced anti-tumor responses compared to either single agent. Several animals in the HT1080 and EBC-1 models that received the combined treatment achieved tumor-free survival (TFS) by study end, or experienced a complete response (CR) or a partial response (PR). Horizontal bars in the upper left indicate dosing period.

IGM-8444 Induces More Potent Anti-Tumor Responses with Birinapant than Anti-DR5 IgG *In Vivo*

MDA-MB-231 Tumor Model

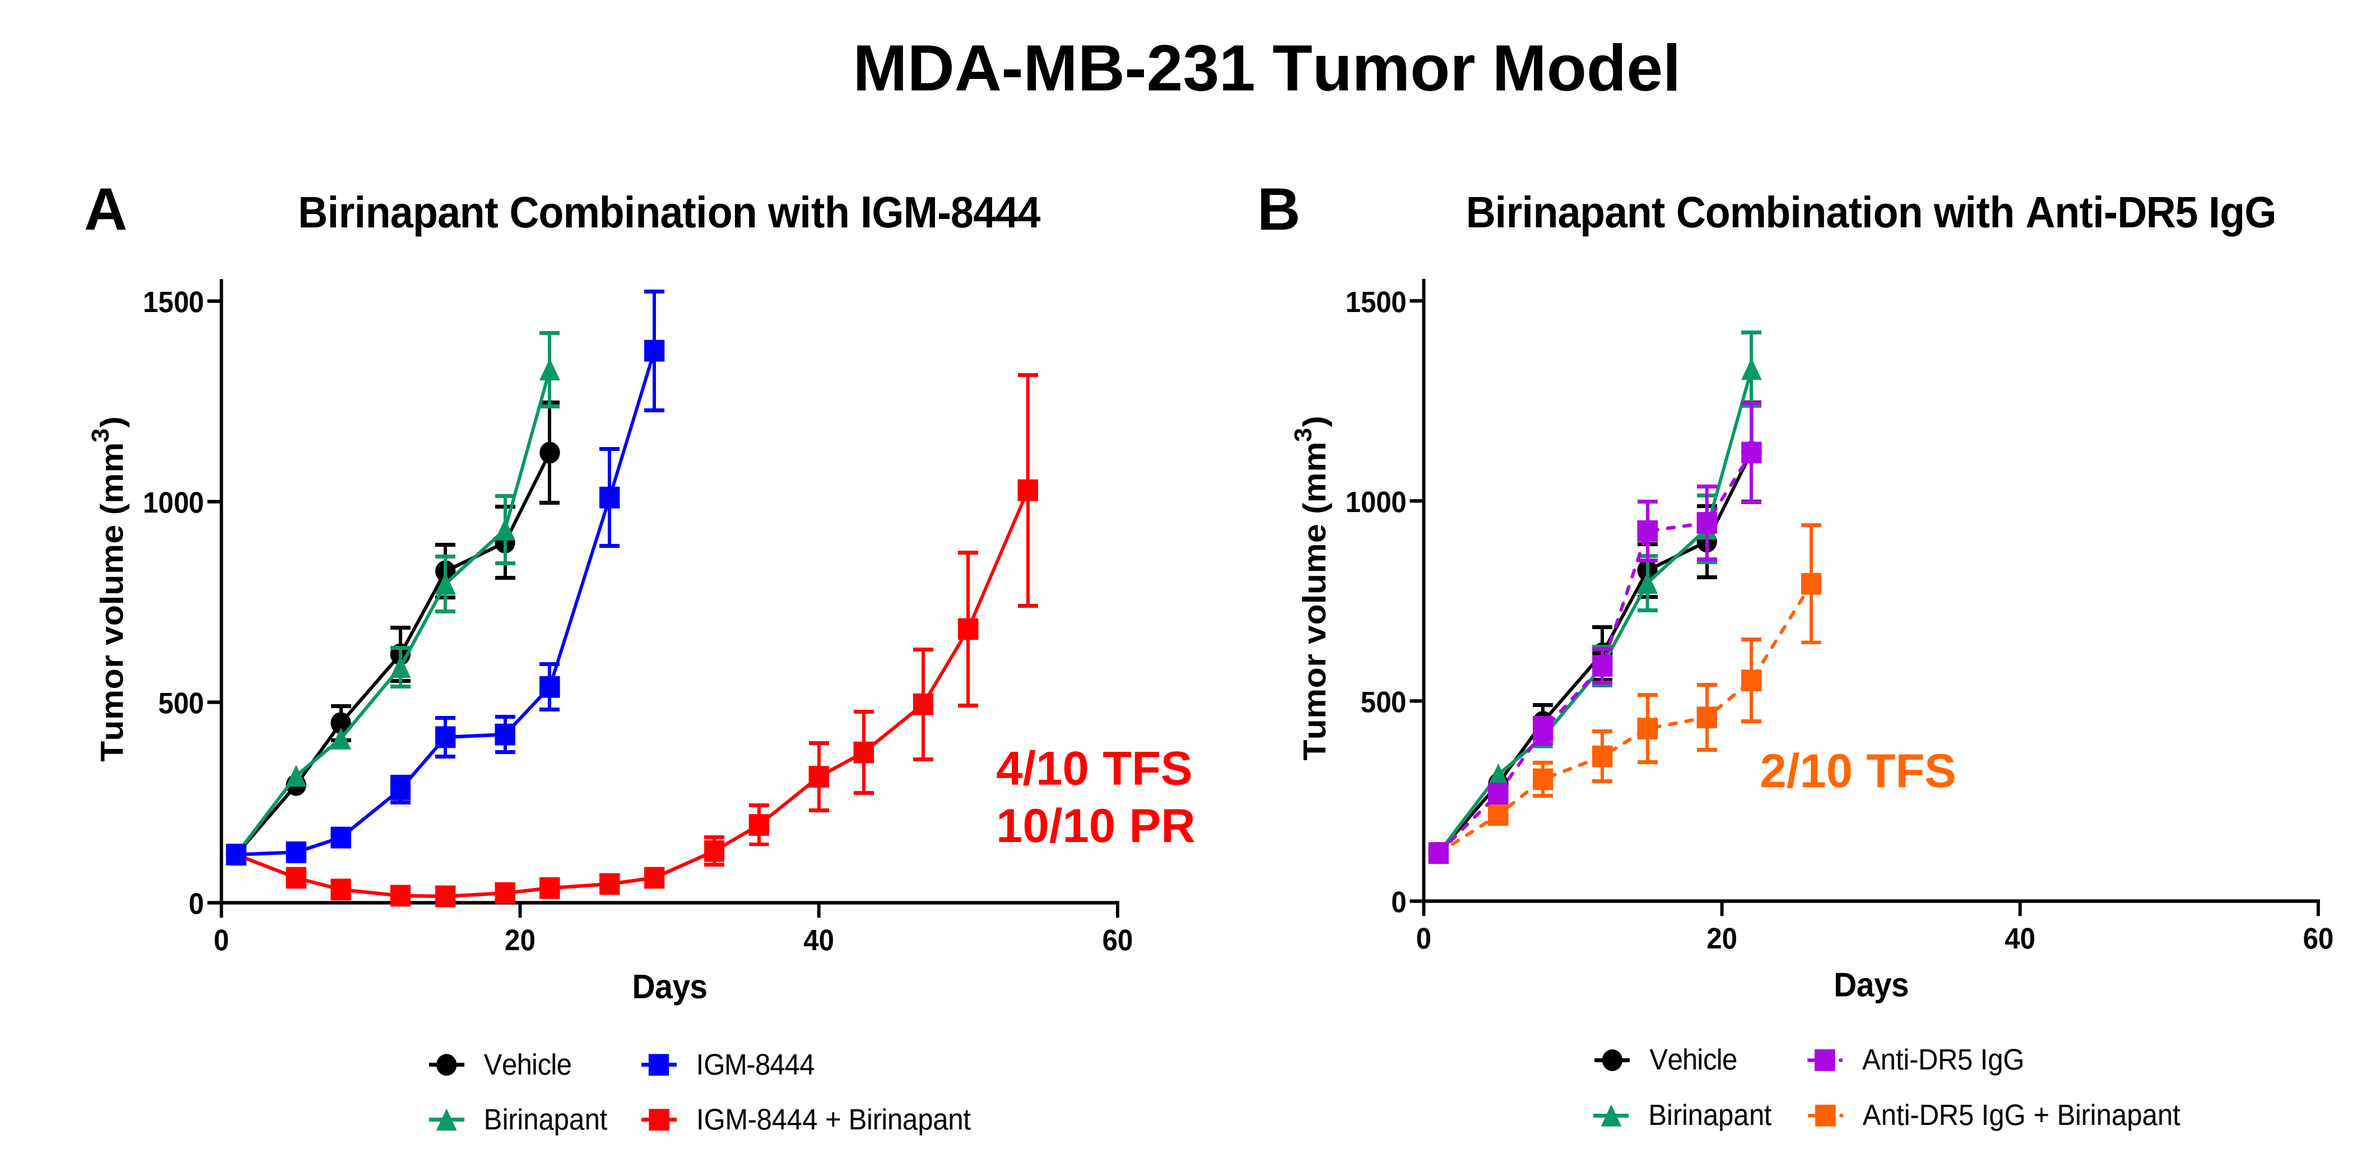

A
Birinapant Combination with IGM-8444
4/10 TFS
10/10 PR
B
Birinapant Combination with Anti-DR5 IgG
2/10 TFS
Tumor volume (mm3)
Days
Vehicle
IGM-8444
Birinapant
IGM-8444 + Birinapant
Anti-DR5 IgG
Anti-DR5 IgG + Birinapant

Figure 6. Comparison of anti-tumor responses between IGM-8444 and an anti-DR5 IgG at roughly matched *in vivo* exposure levels (within 4-fold). Female athymic nude mice were subcutaneously implanted with MDA-MB-231 tumor cells. **A)** Animals were treated with vehicle i.v. q2dx11, IGM-8444 at 5 mg/kg i.v. q2dx11, birinapant at 2.5 mg/kg i.p. q3dx7, or a combination of the IGM-8444 and birinapant dosing regimens. **B)** Animals were treated with vehicle, anti-DR5 IgG at 5 mg/kg i.v. qwx3, birinapant at 2.5 mg/kg i.p. q3dx7, or a combination of the anti-DR5 IgG and birinapant dosing regimens. Despite roughly matched *in vivo* exposure, IGM-8444 showed more anti-tumor responses than anti-DR5 IgG both as monotherapy and in combination with birinapant. TFS, tumor-free survival; PR, partial response.

IGM-8444 and Birinapant Induced Anti-Tumor Responses is Birinapant Dose-Dependent *In Vivo*

MDA-MB-231 Tumor Model

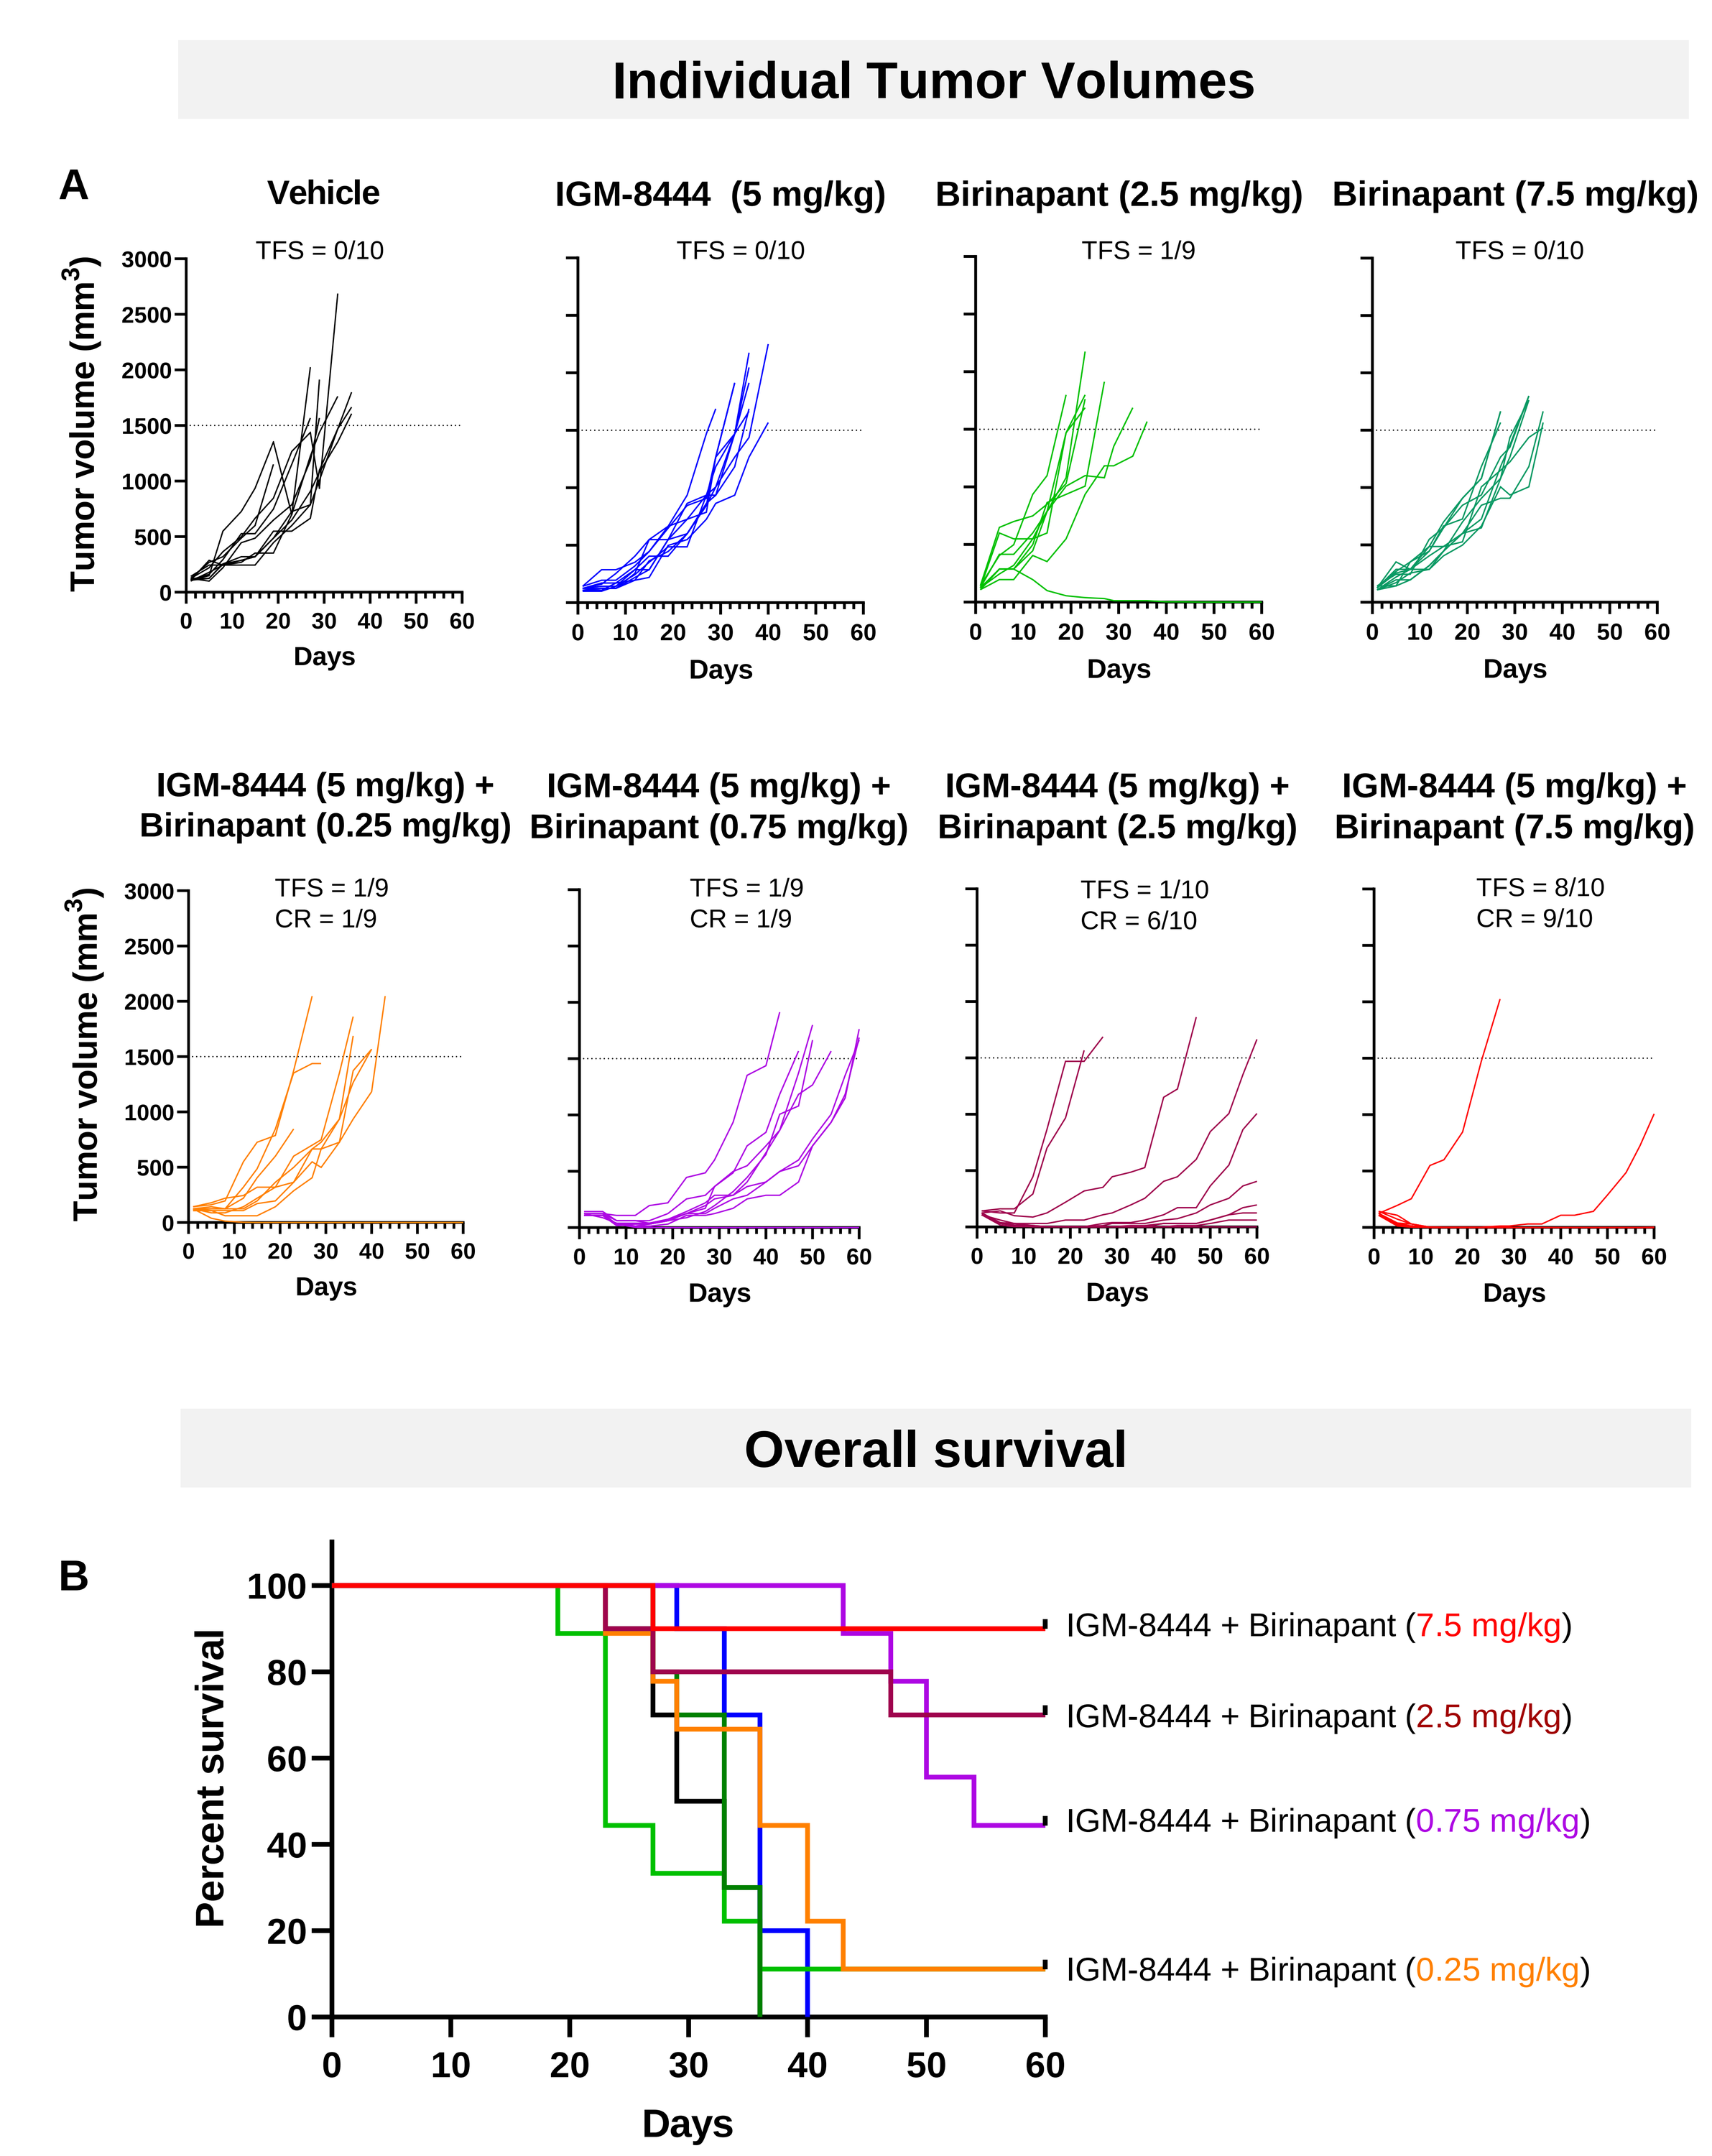

Individual Tumor Volumes
A
Vehicle
TFS = 0/10
IGM-8444 (5 mg/kg)
TFS = 0/10
Birinapant (2.5 mg/kg)
TFS = 1/9
Birinapant (7.5 mg/kg)
TFS = 0/10
IGM-8444 (5 mg/kg) + Birinapant (0.25 mg/kg)
TFS = 1/9
CR = 1/9
IGM-8444 (5 mg/kg) + Birinapant (0.75 mg/kg)
TFS = 1/9
CR = 1/9
IGM-8444 (5 mg/kg) + Birinapant (2.5 mg/kg)
TFS = 1/10
CR = 6/10
IGM-8444 (5 mg/kg) + Birinapant (7.5 mg/kg)
TFS = 8/10
CR = 9/10
Tumor volume (mm3)
Days
Overall survival
B
Percent survival
IGM-8444 + Birinapant (7.5 mg/kg)
IGM-8444 + Birinapant (2.5 mg/kg)
IGM-8444 + Birinapant (0.75 mg/kg)
IGM-8444 + Birinapant (0.25 mg/kg)

Figure 7. Female athymic nude mice were implanted with MDA-MB-231 s.c. When tumors reached 100-150 mm^3, animals were randomized and treated with IGM-8444 at 5 mg/kg i.v. q2dx11, birinapant at 2.5 or 7.5 mg/kg i.p. q3dx7, or with the combination of IGM-8444 fixed dose and birinapant escalating dose from 0.25 mg/kg to 7.5 mg/kg. **A)** Each graph displays the individual tumor volume (mm^3) over time per treatment. The dotted line indicates the tumor volume endpoint, and the number of animals that achieved tumor-free survival (TFS) or a complete response (CR) is indicated above each graph. **B)** The graph represents the survival of animals for each treatment group. Enhanced anti-tumor responses and survival correlated with increasing birinapant dose.

Tissue and Soluble Biomarkers of Apoptosis and IAP Degradation Correlate With Anti-Tumor Responses

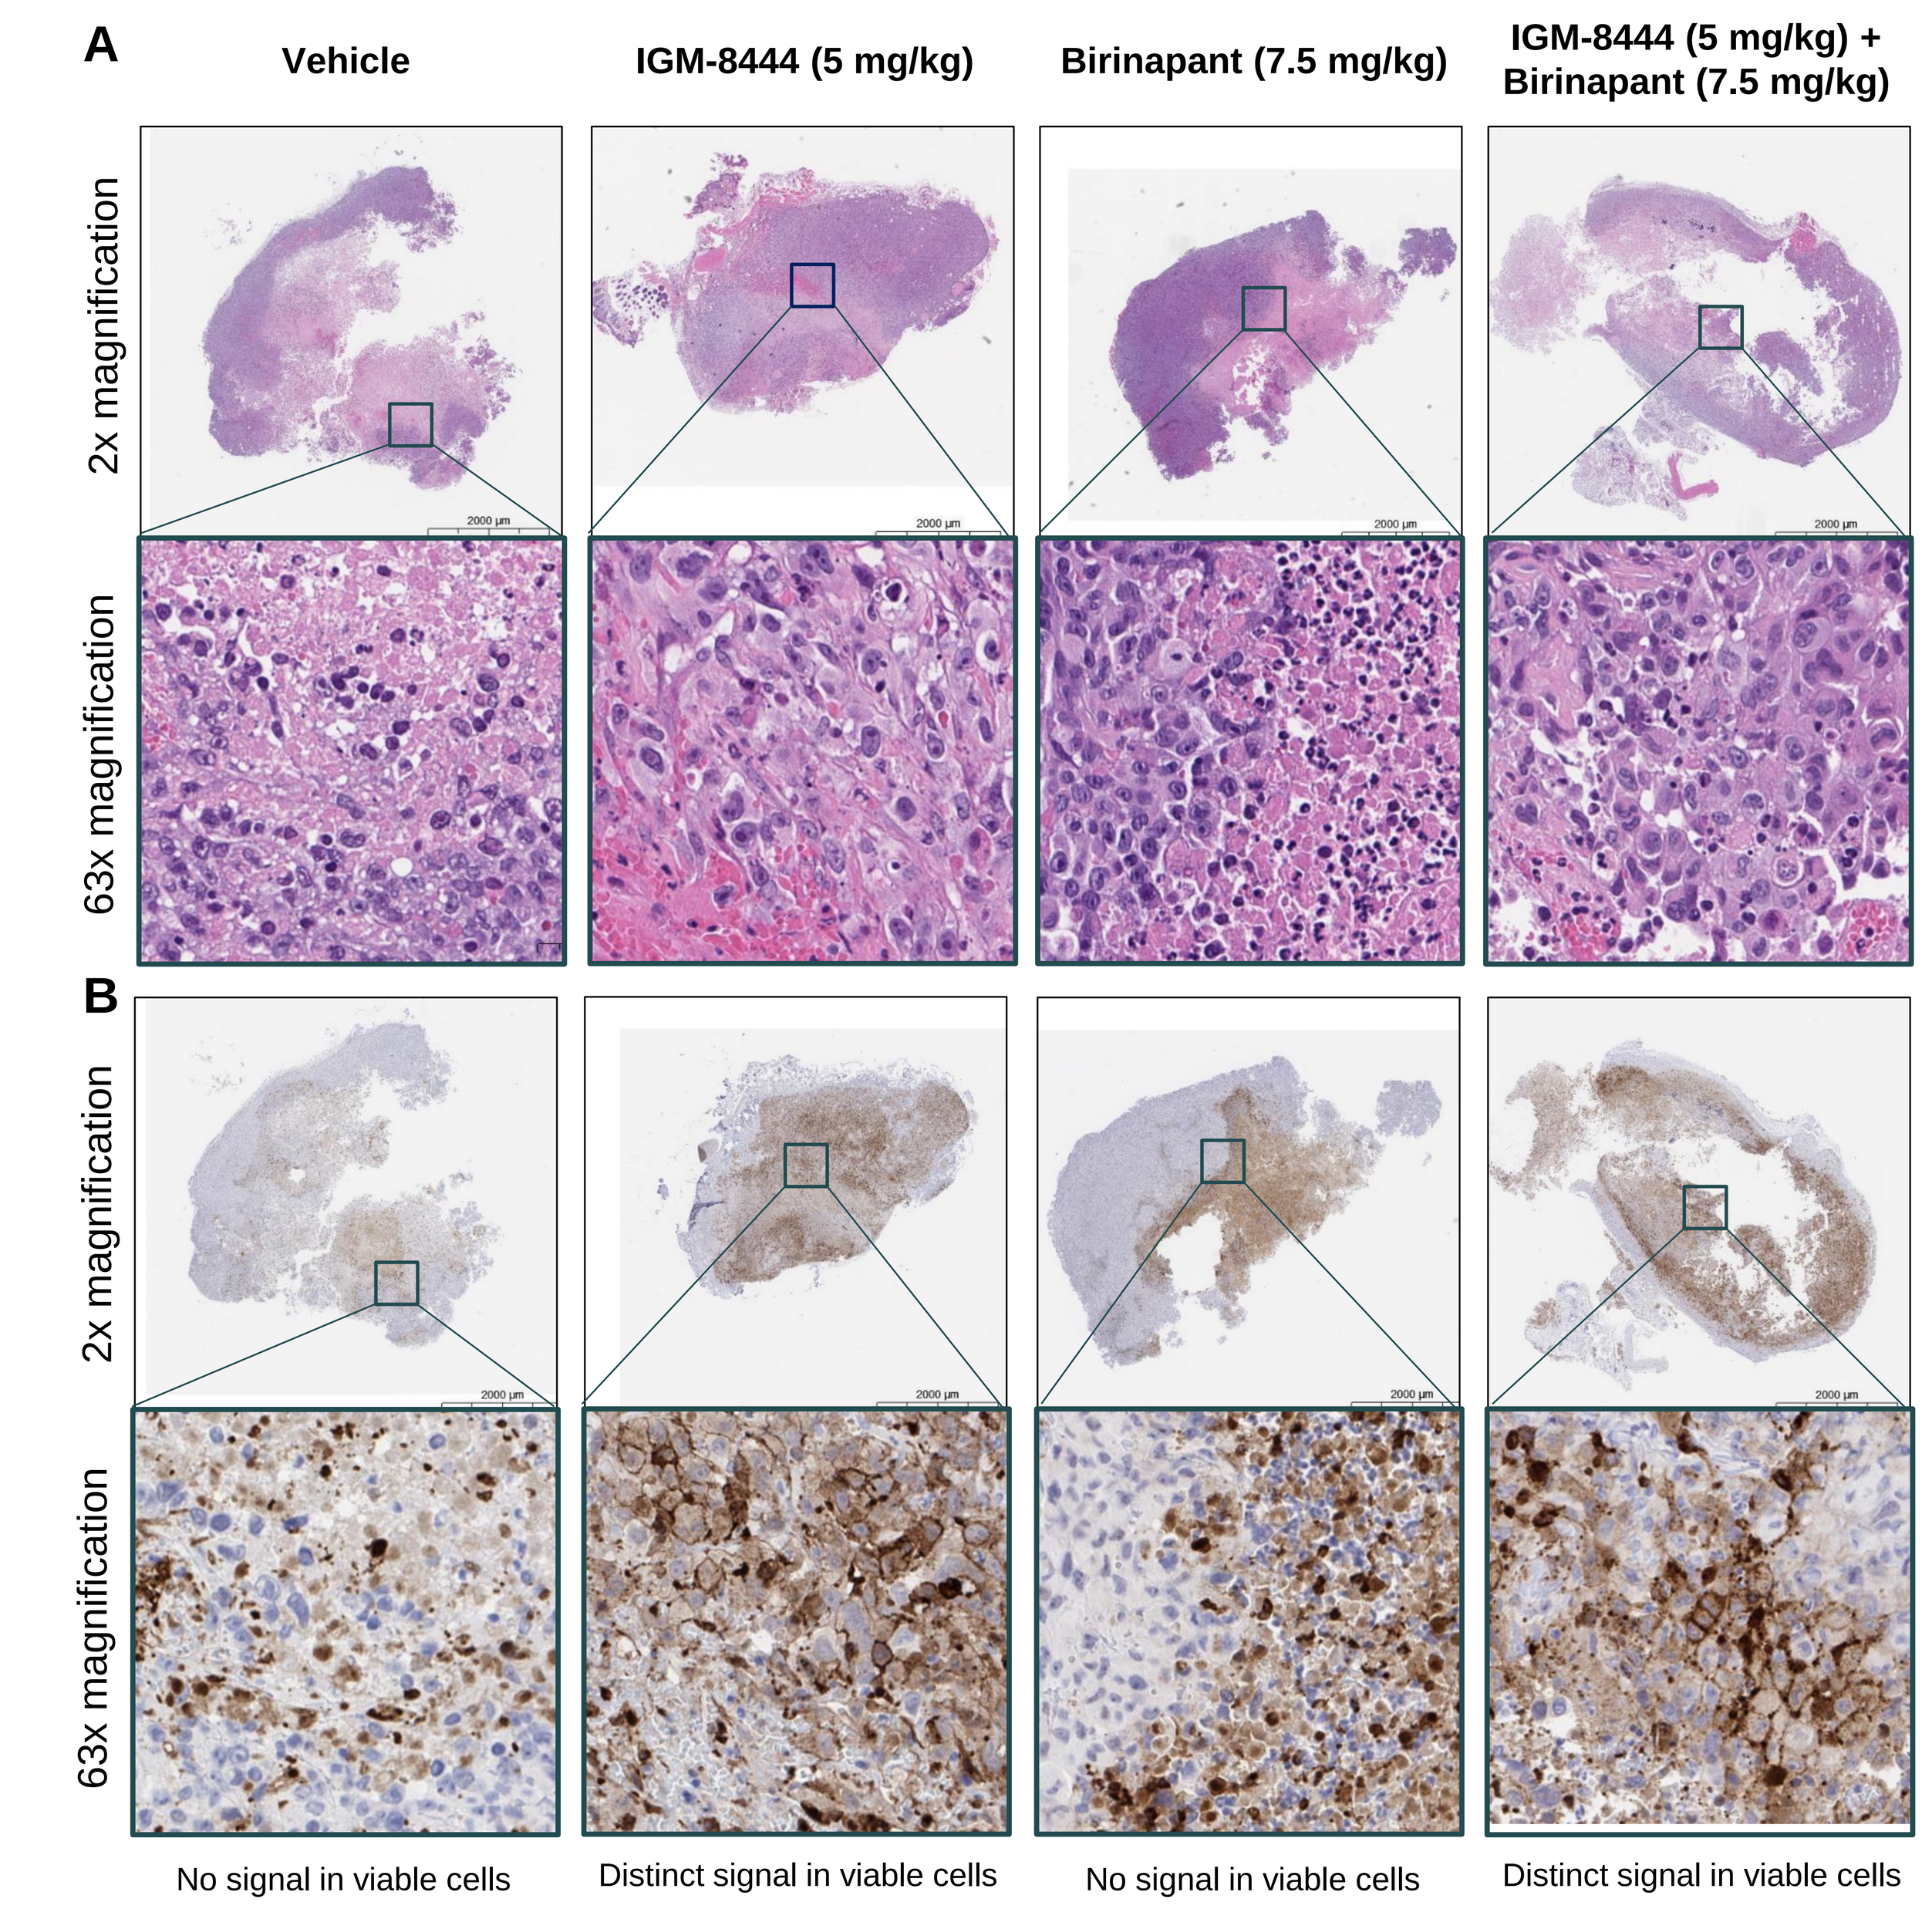

H&E and Tumor Cleaved Caspase 3
A
Vehicle
IGM-8444 (5 mg/kg)
Birinapant (7.5 mg/kg)
IGM-8444 (5 mg/kg) + Birinapant (7.5 mg/kg)
2x magnification
63x magnification
B
No signal in viable cells
Distinct signal in viable cells
No signal in viable cells
Distinct signal in viable cells

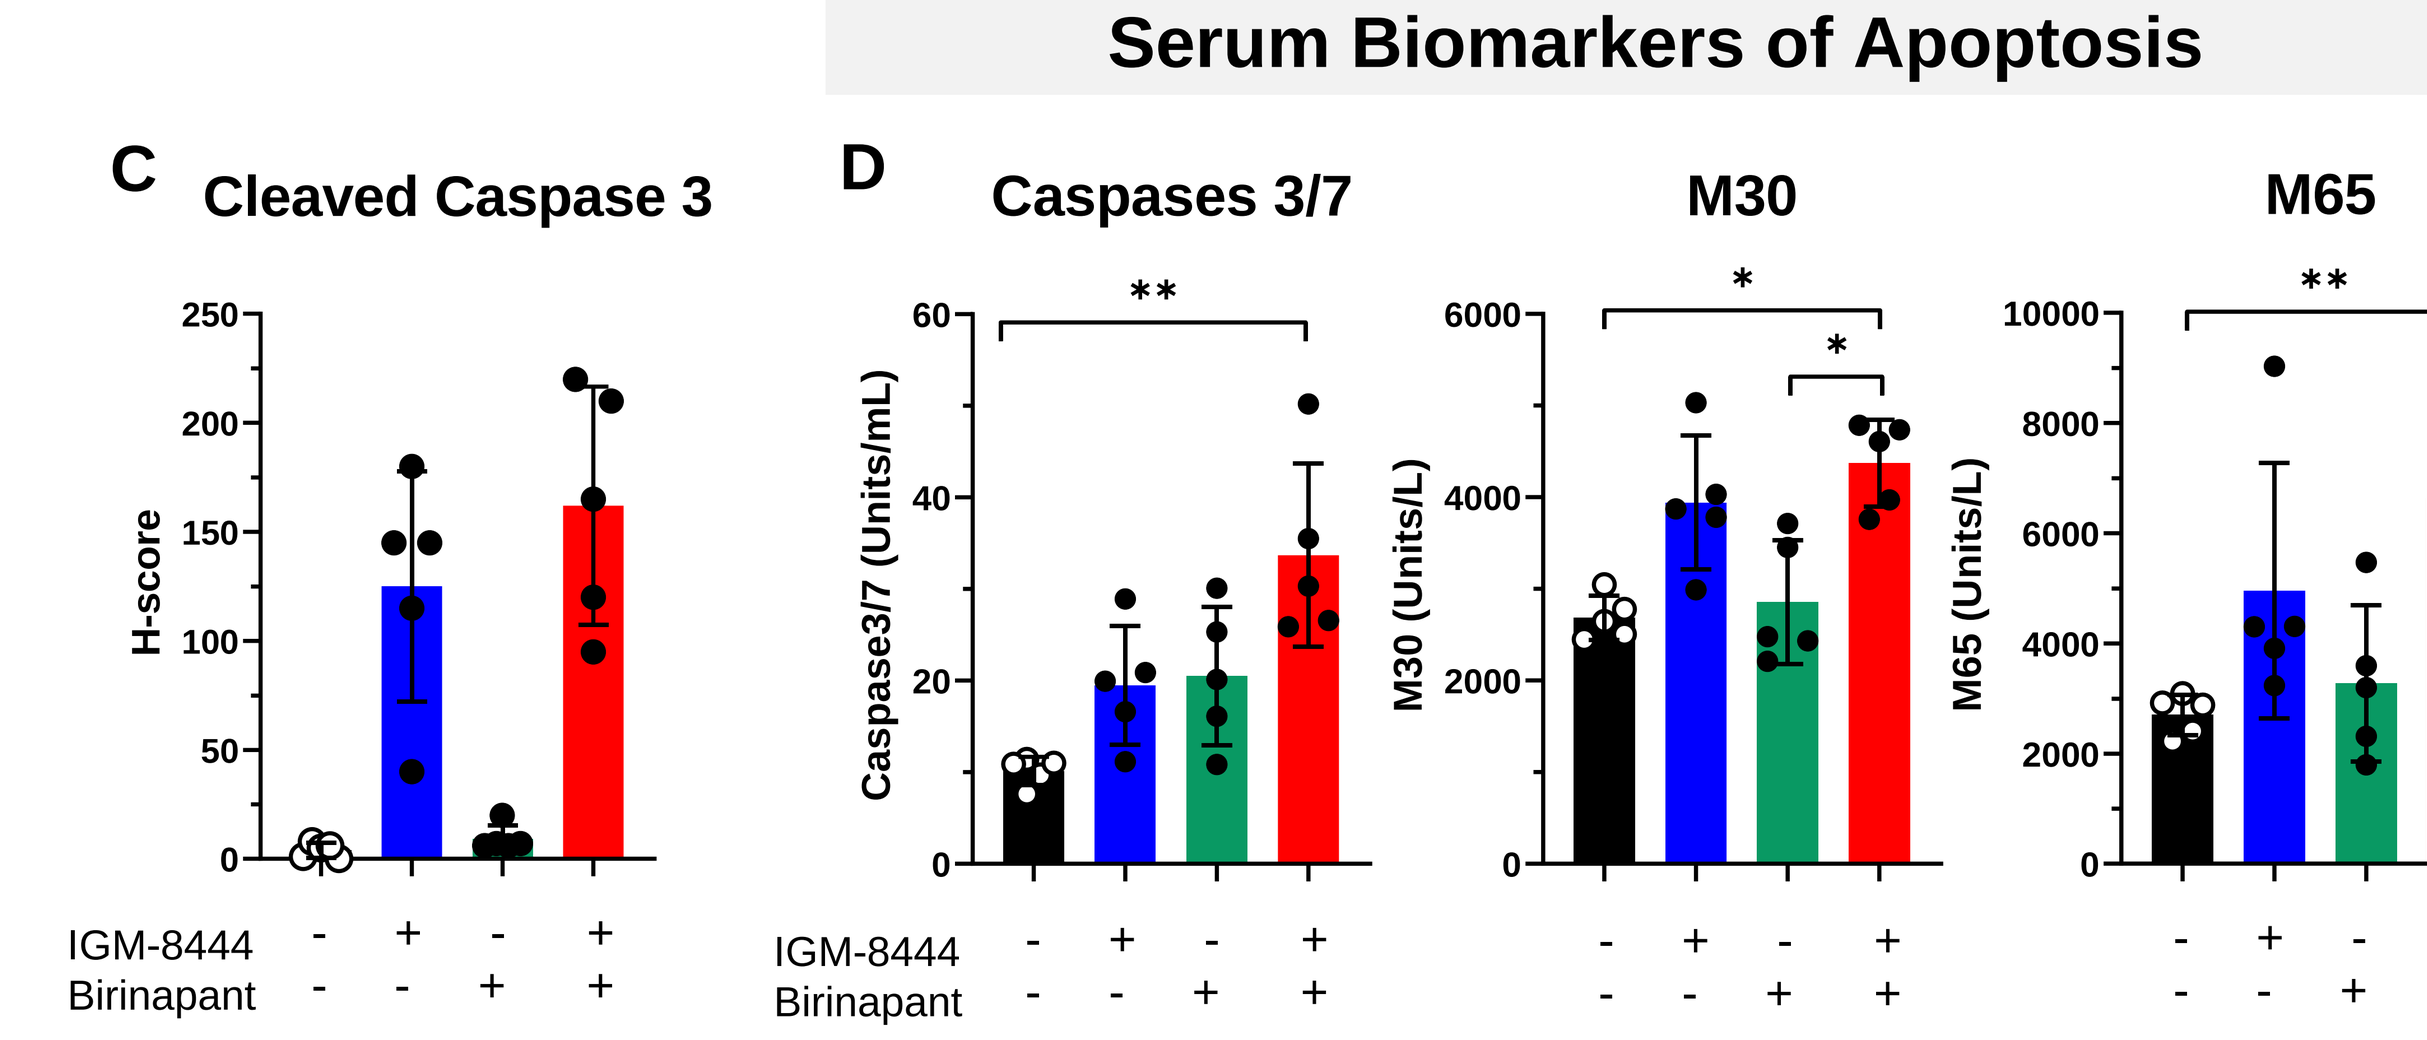

Serum Biomarkers of Apoptosis
C
Cleaved Caspase 3
H-score
D
Caspases 3/7
Caspase3/7 (Units/mL)
M30
M30 (Units/L)
M65
M65 (Units/L)
IGM-8444
Birinapant

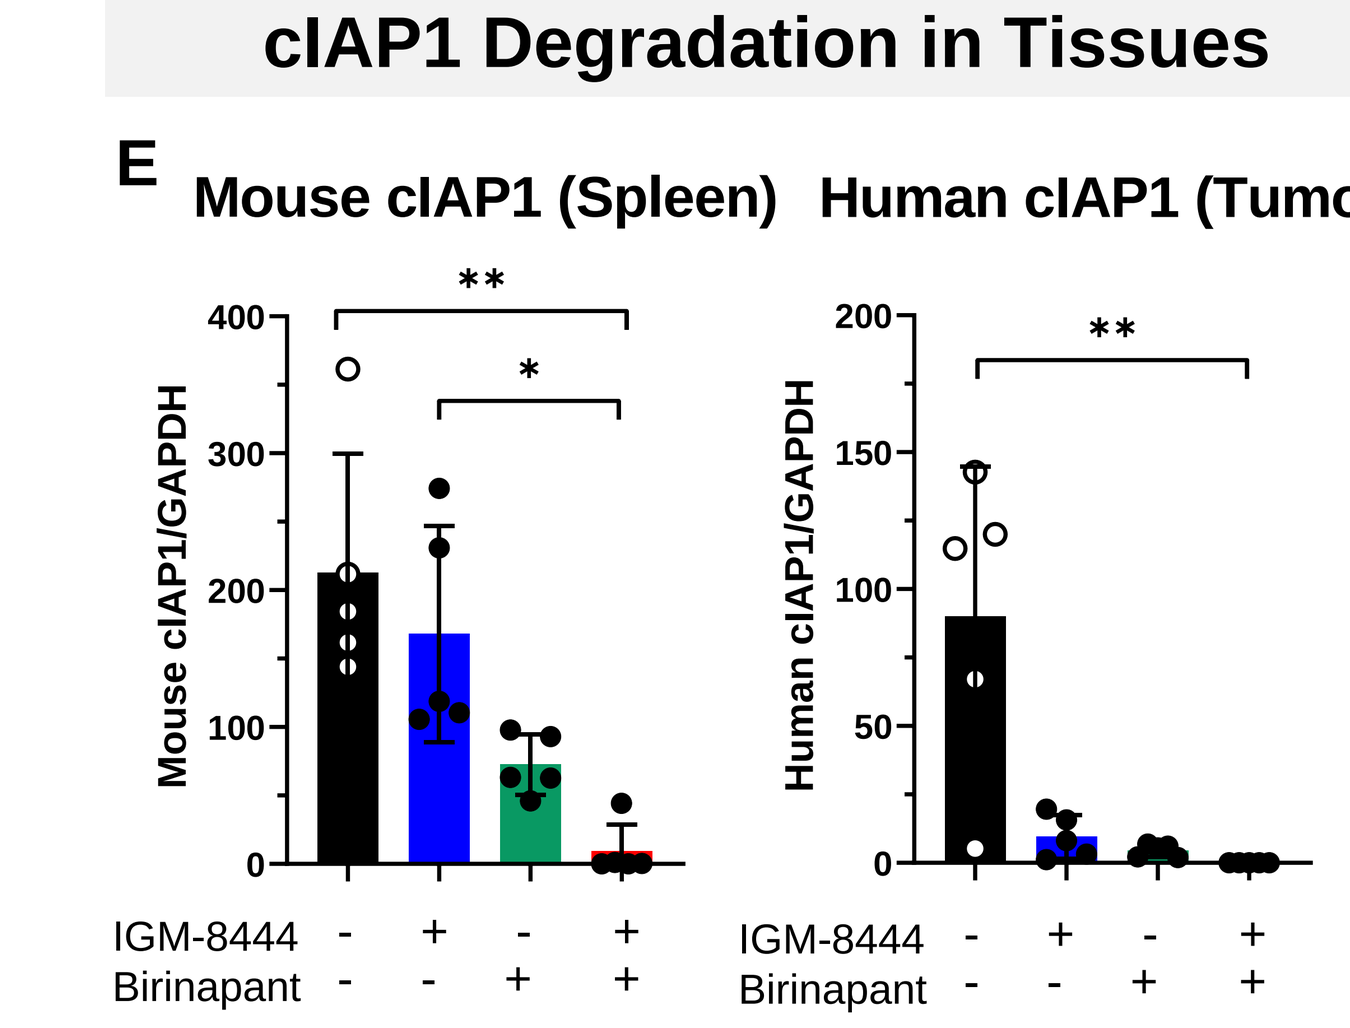

cIAP1 Degradation in Tissues
E
Mouse cIAP1 (Spleen)
Human cIAP1 (Tumor)
Mouse cIAP1/GAPDH
Human cIAP1/GAPDH
IGM-8444
Birinapant

Figure 8. MDA-MB-231 tumor-bearing mice were treated with IGM-8444 (5 mg/kg i.v.) single agent, birinapant (7.5 mg/kg i.p.) single agent or both compounds. Twenty-four hours after the first dose serum or tissues (spleen, tumor) were collected. Representative images of H&E staining **(A)** and cleaved caspase 3 IHC **(B)** in all 4 groups with 2X and 63X magnifications. **C)** H-scores of cleaved caspase 3 signals from viable cells. **D)** Serum biomarkers (caspase 3/7, M30 and M65) were analyzed by ELISA. **E)** Mouse cIAP1 and human cIAP1 were analyzed by western blot on tissue homogenates from spleen or tumor, respectively. The expression was normalized to GAPDH. Mean ±SD is shown. Statistical significance is calculated with a Kruskal-Wallis test followed by a Dunn's test (*p<0.05, **p<0.01). Changes in pharmacodynamic biomarkers were consistent with IGM-8444 and birinapant mechanisms of action.

Summary

- IGM-8444 and birinapant combination induced synergistic cytotoxicity in 36/45 (80%) cancer cell lines screened, without killing primary human hepatocytes *in vitro*
- IGM-8444 combination with birinapant sensitized cells that developed acquired resistance to DR5 agonist antibodies
- IGM-8444 and birinapant induced tumor regressions in several models, and showed dose-dependent anti-tumor responses with increasing birinapant dose in a TNBC model; by comparison, birinapant combination with an anti-DR5 IgG showed only a modest response
- Pharmacodynamic biomarkers including caspase activation and cIAP1 degradation correlated with anti-tumor response
- The combination of IGM-8444 with birinapant is currently under evaluation in a Phase 1 study in patients with relapsed and/or refractory cancers (NCT04553692)

Poster No. 1068, 2022 AACR Meeting, New Orleans, April 8-13

https://igmbio.com